P1601

Melanocytes' dendricity down-regulated by the association niacinamide - ascorbic acid

Marius A. Ionescu, MD, PhD[1,2], Luc Lefeuvre[2], Anne-Marie Matta[2], Michel Bohbot, PhD[2], Agnès Gougerot, MD[2]

1. Saint-Louis Hospital, Paris; 2. Laboratoires Dermatologiques d'Uriage, Courbevoie, France

Background

Melanocytes activated by solar radiation increase their dendricity (melanin transfer) and melanin synthesis, involving the expression of tyrosinase family of proteins including tyrosinase-related protein 1 and 2 (TRP1, TRP2)[1,2].

Purpose

To assess the effects of the association niacinamide-ascorbic acid on melanogenesis process in human skin explants exposed to solar simulated radiation (SSR).

Methods

Normal human skin explants were treated (untreated control) by an O/W emulsion based on the association niacinamide - ascorbic acid at 2 mg / cm2 of explant, once per day from baseline (D0) to day 9, thirty minutes before an exposure to Solar Simulated Radiation SSR (Vilbert Lourmat® RMX 3W, visible light + UVA+UVB, dose 1.125 J/cm2). After treatment and SSR exposure, explants were studied at baseline, D5 and D12. General morphology of the skin was assessed by optical microscopy (Masson–Goldner); melanin was visualized on silver staining (Masson – Fontana); immunohistochemistry was used for labeling of tyrosinase-related protein-1 (TRP1)(antibodies anti-TRP-1, MEL 5, Calbiochem®).

Results

Thirty nine explants were studied. There were no morphology changes in treated/irradiated explants compared to control. In untreated irradiated explants, at D5 and D12, the melanocytes' dendricity and melanin transfer were induced or increased (Fig 1B and 2B) compared to control (Fig 1A)($p<0.01$). In treated and irradiated explants (Fig 1C and 2C), melanocytes dendricity and melanin transfer were significantly less important compared to untreated, irradiated explants (2B). TRP-1 expression was the same in all explants.

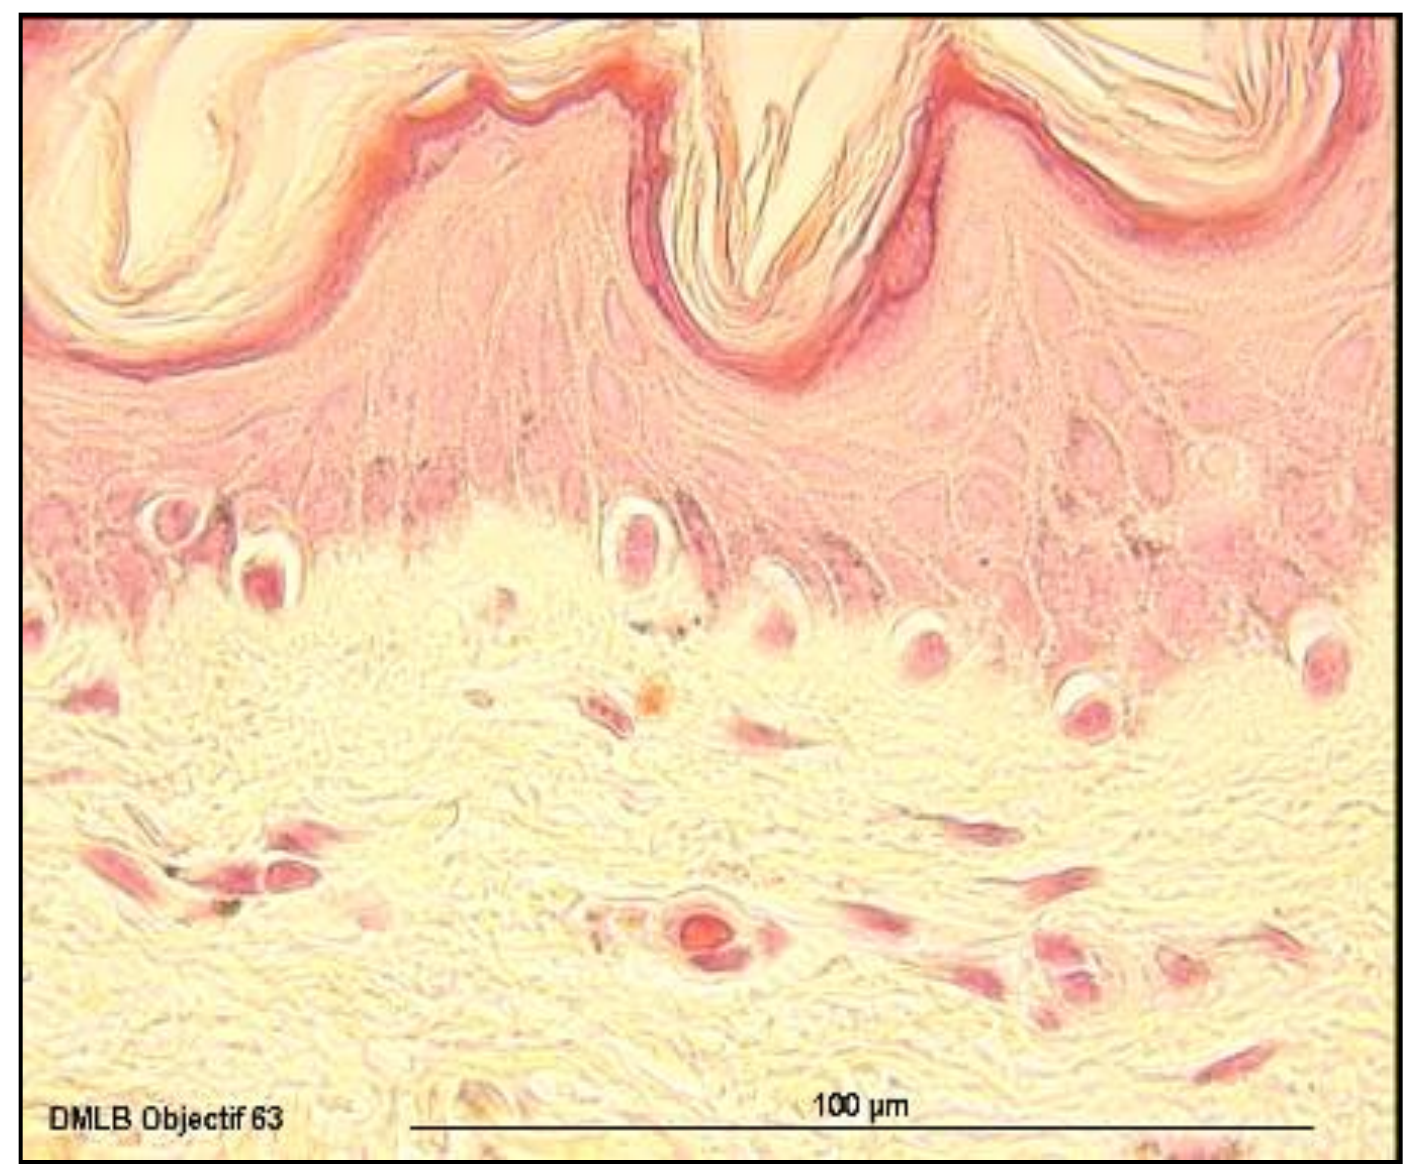
DMLB Objectif 63
100 µm

Fig. 1.A. Control explant at baseline D0
(silver Masson / Fontana)

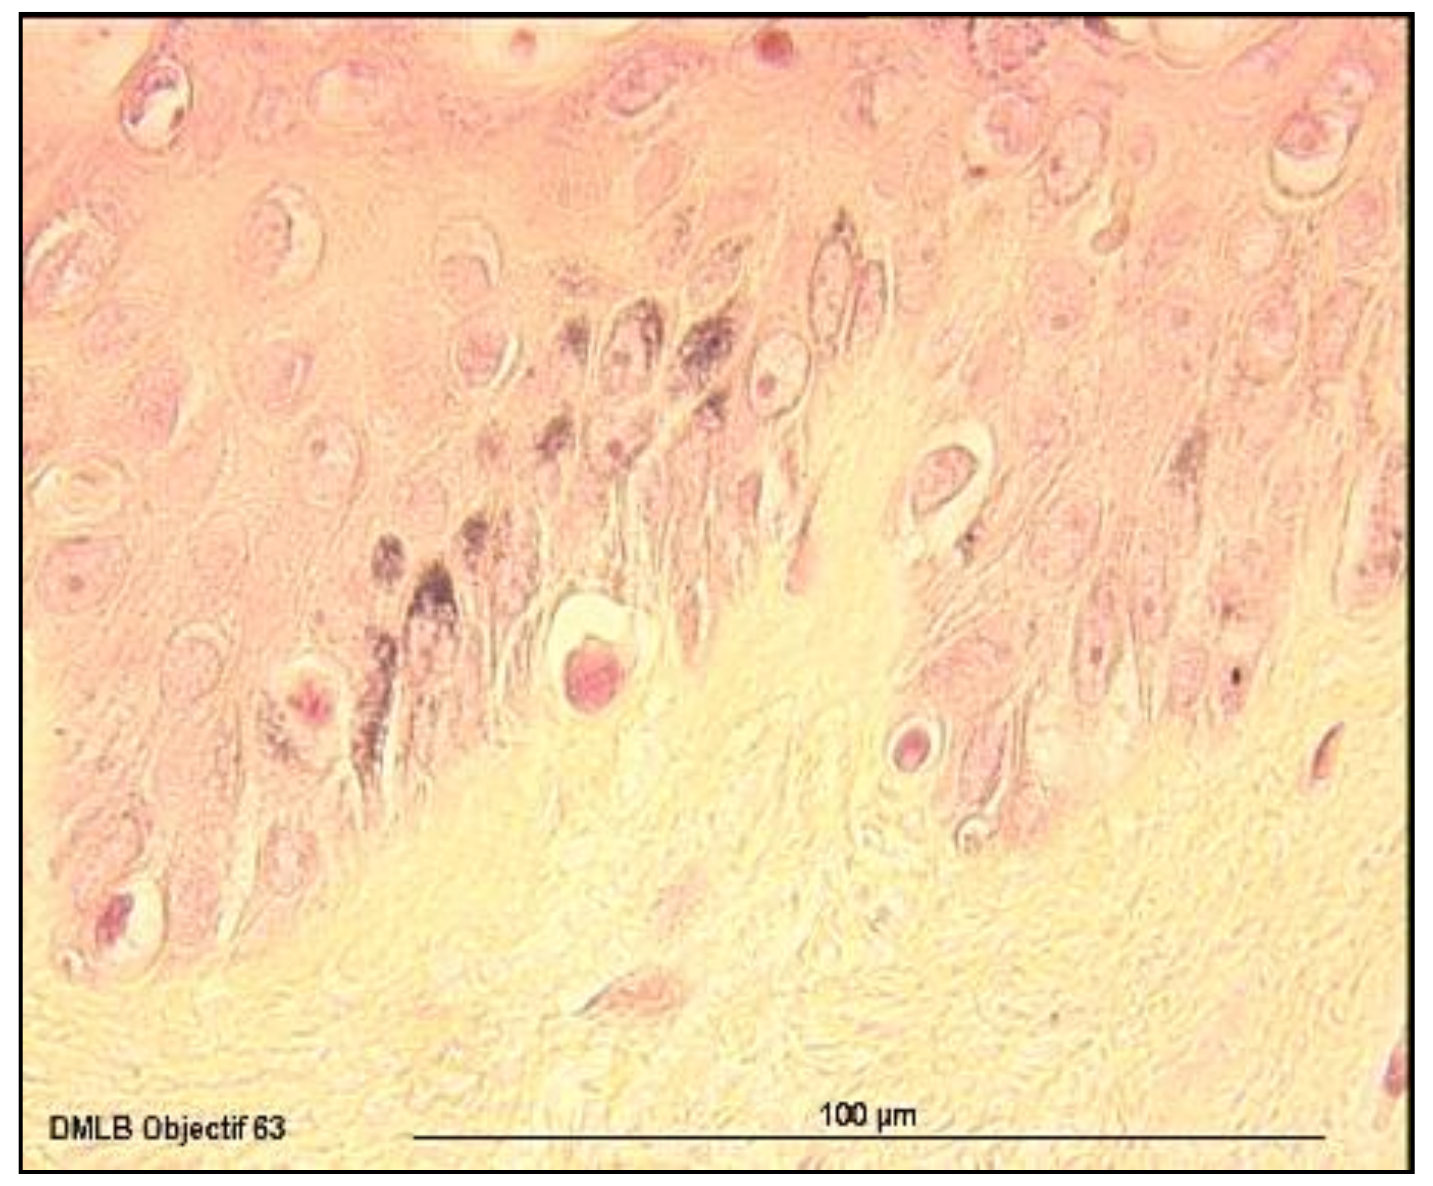
DMLB Objectif 63
100 µm

Fig. 1.B. Untreated, SSR irradiated explant at D12
(silver Masson / Fontana)

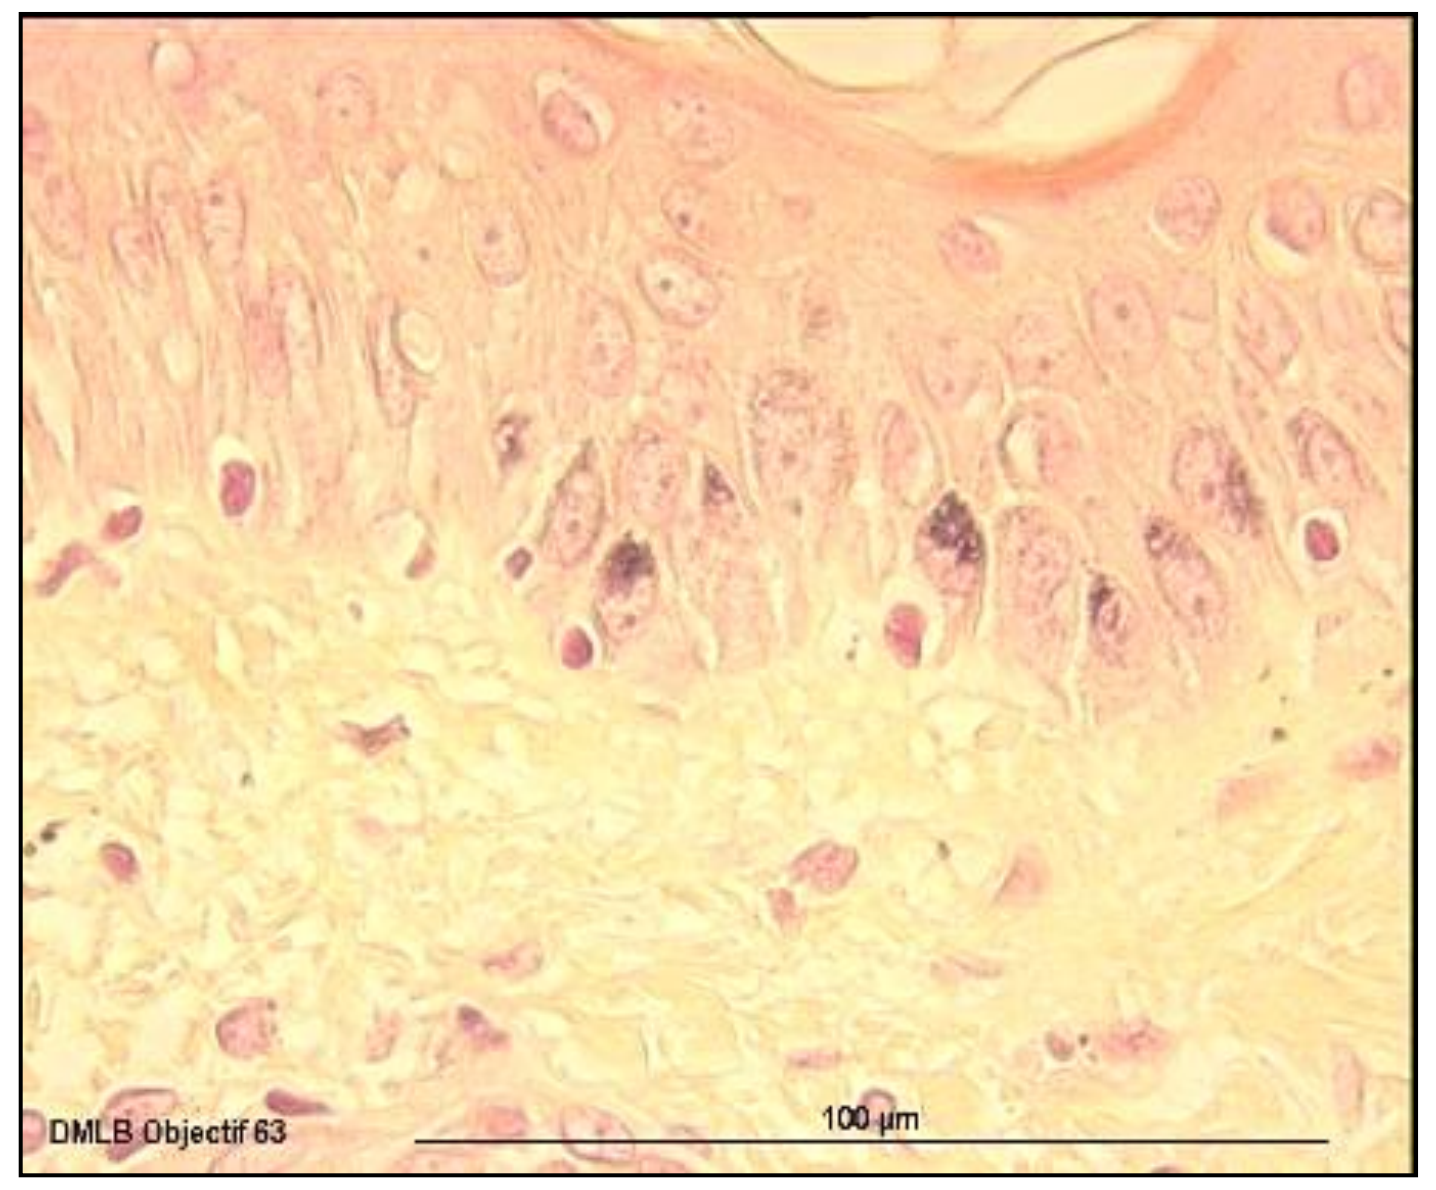
DMLB Objectif 63
100 µm

Fig. 1.C. Treated, SSR irradiated explant at D12
(silver Masson / Fontana)

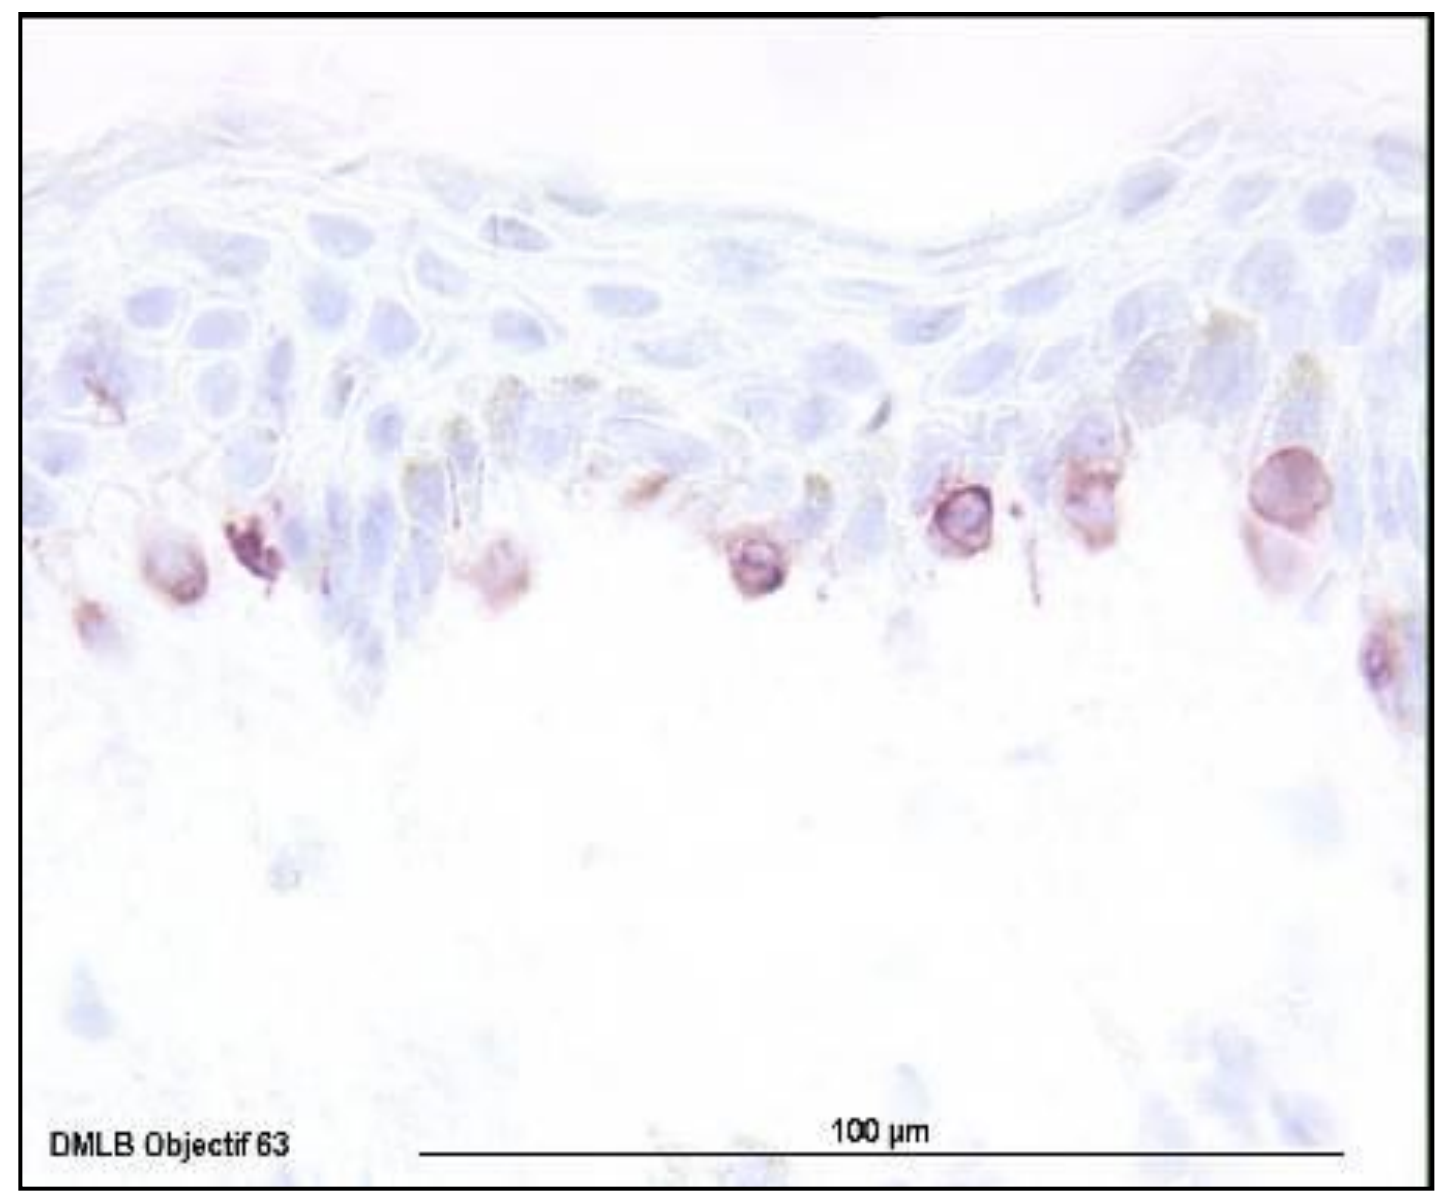
DMLB Objectif 63
100 µm

Fig 2.A. Control explant at baseline D0
(immunohistochemistry MAB anti TRP-1)

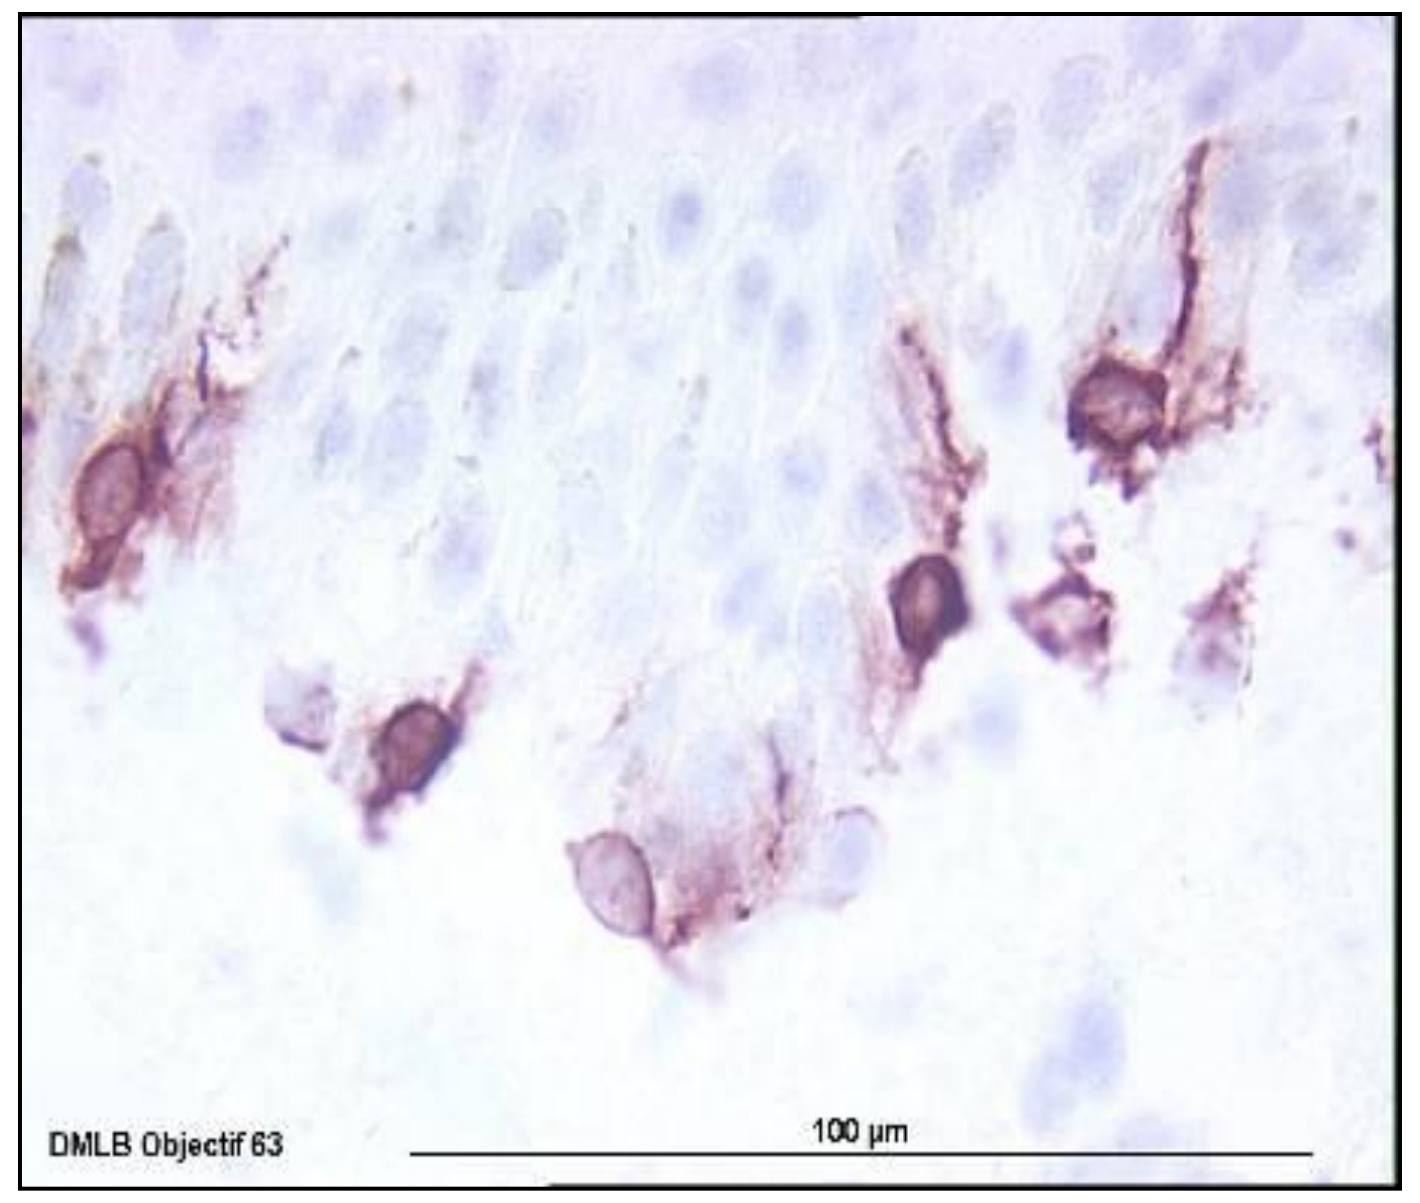
DMLB Objectif 63
100 µm

Fig 2.B. Untreated, SSR irradiated explant at D12
(immunohistochemistry MAB anti TRP-1)

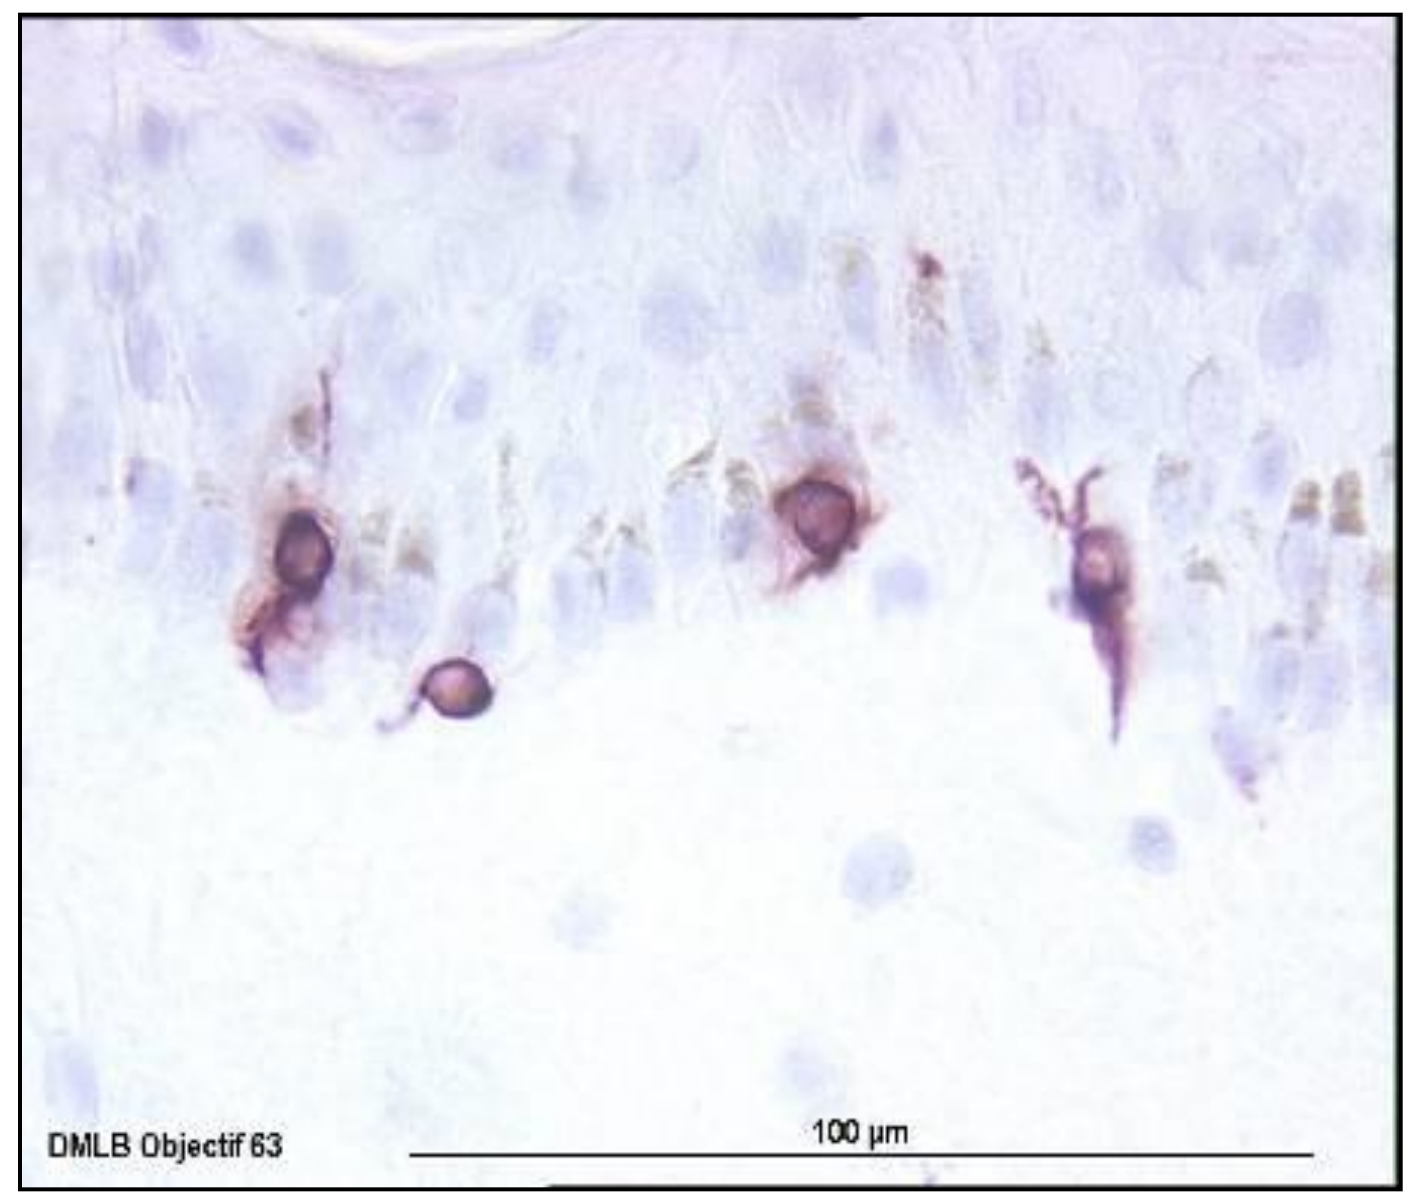
DMLB Objectif 63
100 µm

Fig. 2.C. Treated, SSR irradiated explant at D12
(immunohistochemistry MAB anti TRP-1)

Conclusions

In normal human skin explants treated by the *association* niacinamide-ascorbic acid, then submitted to solar simulated radiation, melanocytes' dendricity and melanin transfer were significantly less important compared to untreated, irradiated explants ($p<0.01$).

References

1. **Huang HY, et al.** ***J Dermatol Sci 2008;49(2):115-23.***
2. **Aoki H, et al.** ***Br J Dermatol 2007;156(6):1214-23.***

American Academy of Dermatology – San Francisco 2009

Supported 100% by Laboratoires Dermatologiques d'Uriage®, France